The Oral Eosinophil-lowering Drug Dexpramipexole Improves FEV_1 Largely through its Effect on FVC

#052

C. Prussin[1], M. E. Bozik[1], J.L. Mather[1], D. G. Archibald[1], S. I. Dworetzky[1], R. Killingsworth[1], S.I. Ochkur[2], E.A. Jacobsen[2], R. Sorkness[3], S. Siddiqui[4], W.W. Busse[5]

1. Knopp Biosciences, Pittsburgh, PA
2. Mayo Clinic Arizona, Scottsdale, AZ
3. University of Wisconsin School of Pharmacy, Madison, WI,
4. National Heart and Lung Institute, Imperial College London, UK.
5. University of Wisconsin School of Medicine and Public Health, Madison, WI,

Introduction & Objectives

Dexpramipexole is an oral investigational drug that lowers blood and tissue eosinophils by inhibiting eosinophil maturation. EXHALE was a dose-ranging RDBPC Phase 2 trial that enrolled 103 moderate-to-severe eosinophilic asthma subjects with absolute eosinophil count (AEC) ≥300/μL, FEV_1 <80% predicted and ≥12% reversibility. Subjects received dexpramipexole 37.5, 75, or 150 mg, or placebo BID for 12 weeks.

Airflow obstruction in asthma (measured by FEV_1) can be partitioned into components of air trapping in small airways (measured by FVC) and airway narrowing (measured by FEV_1/FVC).[1] Anti-eosinophil treatments improve lung function in asthma, but the effect of eosinophil depletion on individual components of airflow obstruction, i.e. FEV_1/FVC vs FVC, are not established and may provide insight into the effects of eosinophils on pulmonary physiology. To that end, we measured the percentage change in FVC and FEV_1/FVC ratio during the EXHALE trial to understand the relative contribution of of air trapping and airway narrowing to improvements in FEV_1.

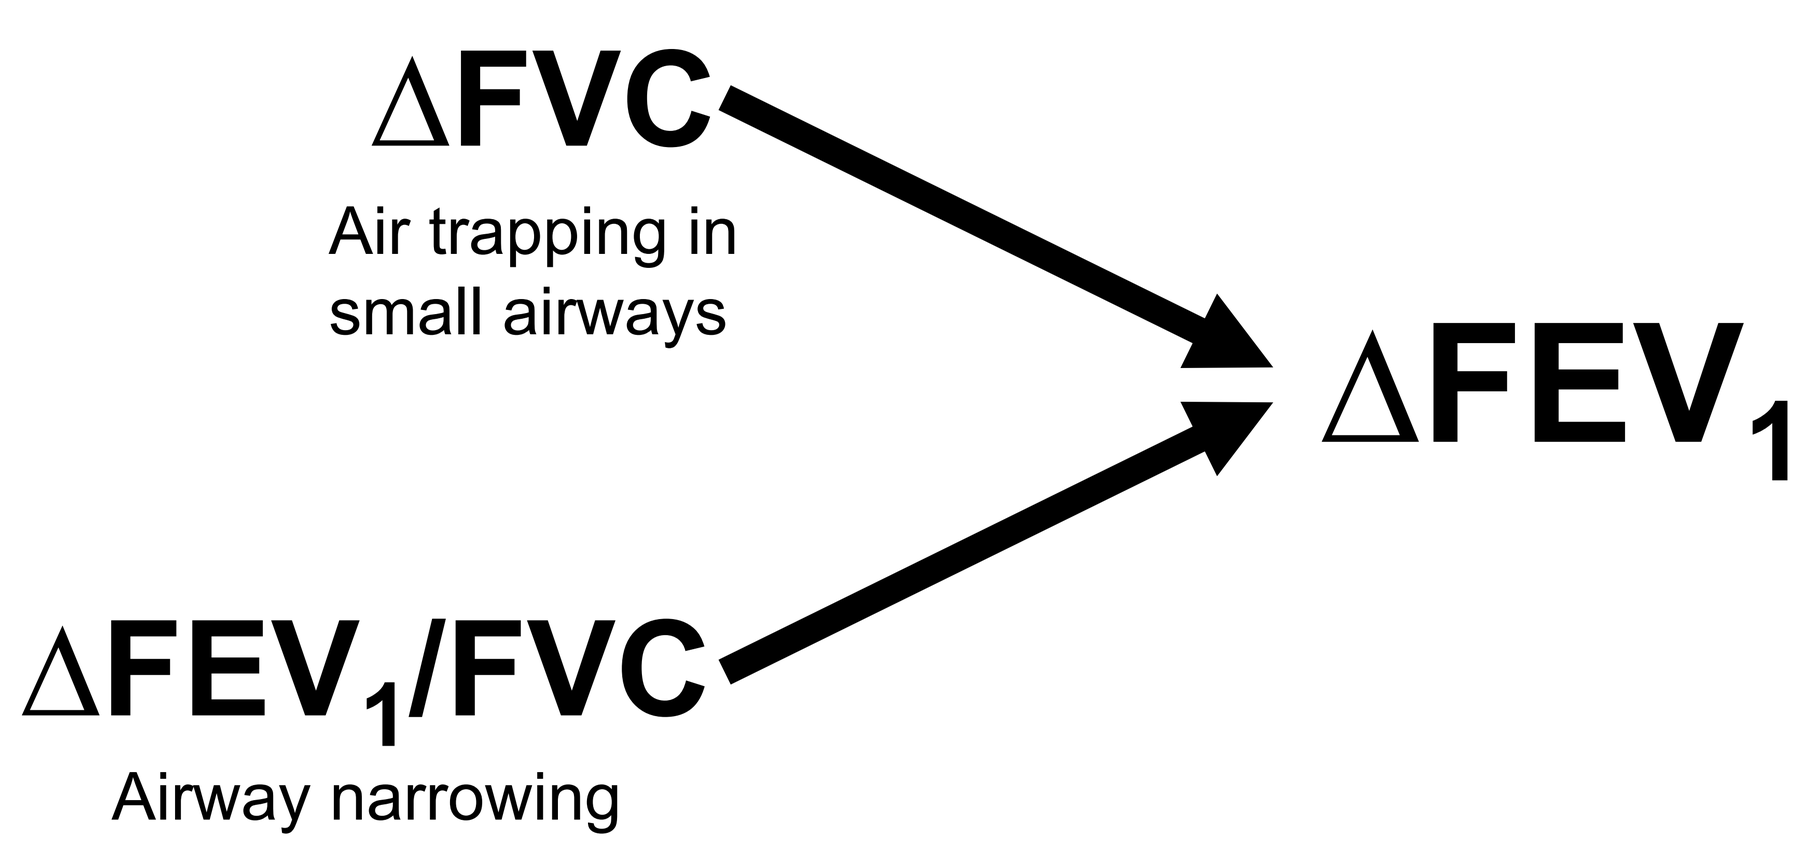

ΔFVC
Air trapping in small airways
ΔFEV1
ΔFEV1/FVC
Airway narrowing

Study Design/Methods

• EXHALE was a randomized, double-blind, placebo-controlled trial of dexpramipexole in eosinophilic asthma, NCT04046939 (Figure 1).

Key inclusion criteria:
• Age 18-75 years, GINA steps 3-5, on daily inhaled corticosteroid plus long-acting beta agonist.
• Pre-bronchodilator FEV_1 <80% & ≥40% predicted, with reversibility ≥12% and ≥200 mL (at Screening).
• Absolute eosinophil count (AEC) ≥300/μL (at Screening).
• Asthma control questionnaire-7 (ACQ-7) ≥1.5 (at Screening).

Endpoints:
Primary Endpoint
• Change in blood absolute eosinophil count from baseline (BL) to Week 12 (W12)
Secondary & Exploratory Endpoints
• Change in pre-bronchodilator (pre-BD) FEV_1, from BL to Week 12
• Change in nasal eosinophil peroxidase (EPX) to protein ratio. Nasal EPX is correlated to sputum eosinophil count and has been identified as a surrogate for lower airway eosinophils.[2]

Fig. 1 EXHALE study design

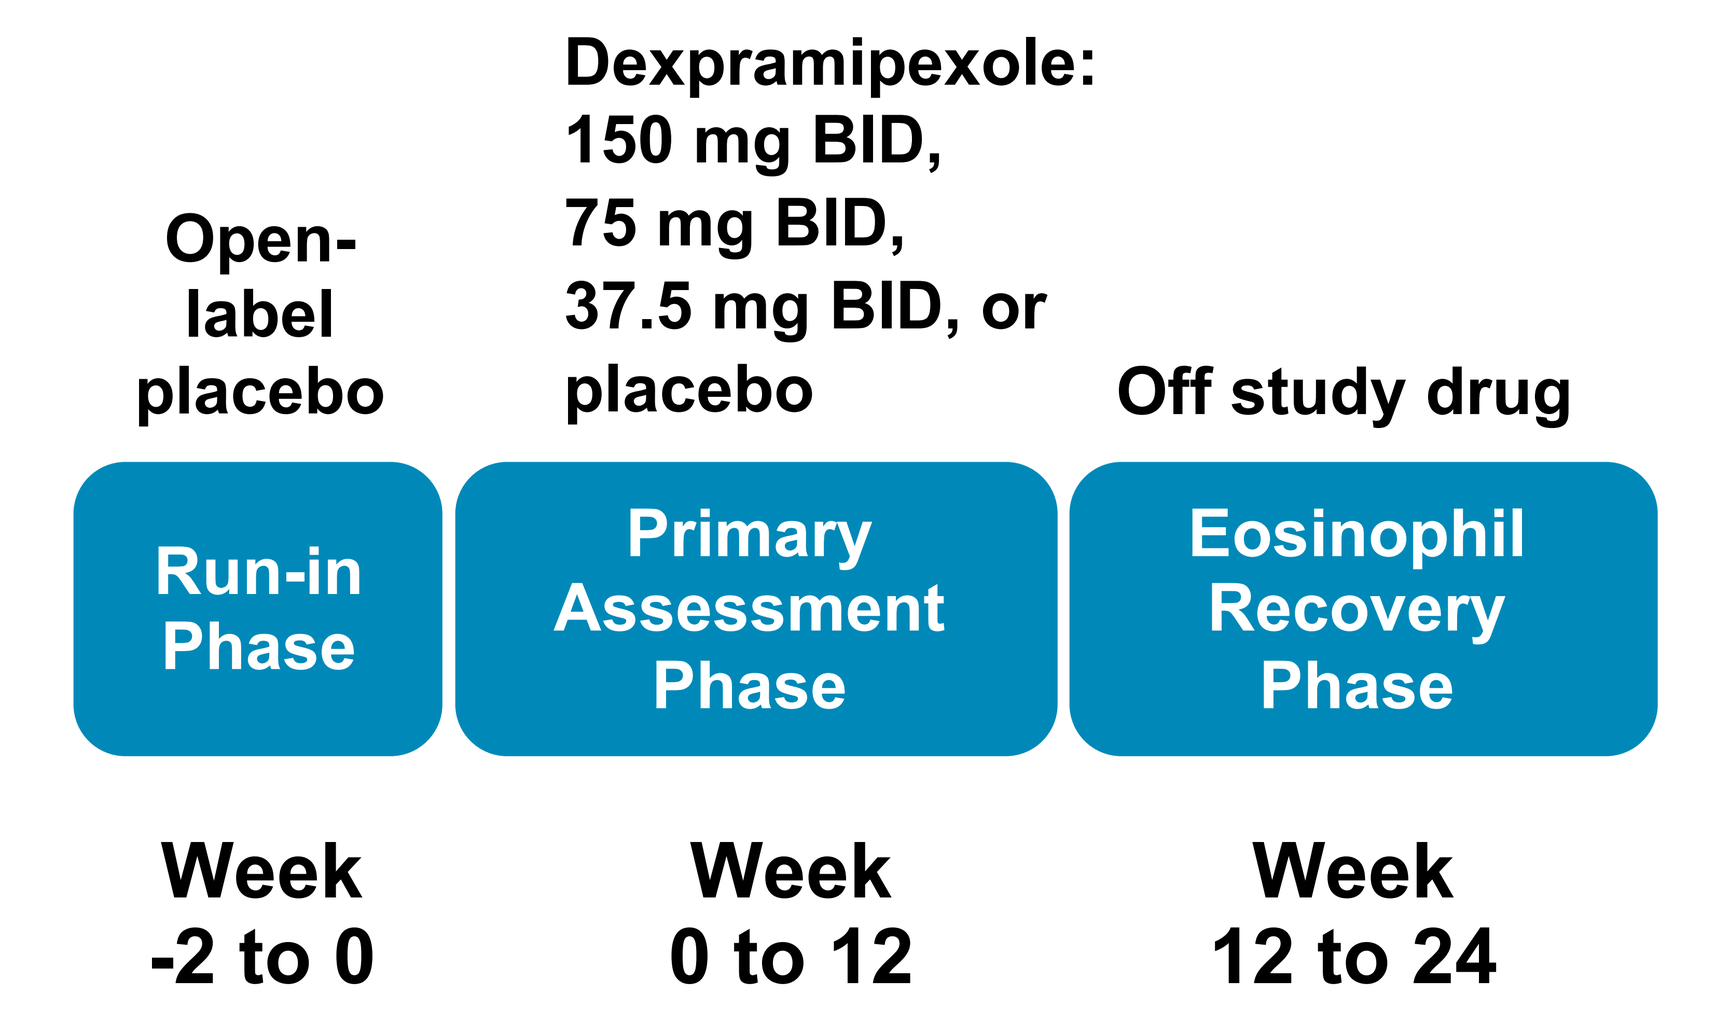

Open-label placebo
Dexpramipexole: 150 mg BID, 75 mg BID, 37.5 mg BID, or placebo
Off study drug
Run-in Phase
Primary Assessment Phase
Eosinophil Recovery Phase
Week -2 to 0
Week 0 to 12
Week 12 to 24

Results

Fig. 2 Primary endpoint, change in eosinophil count

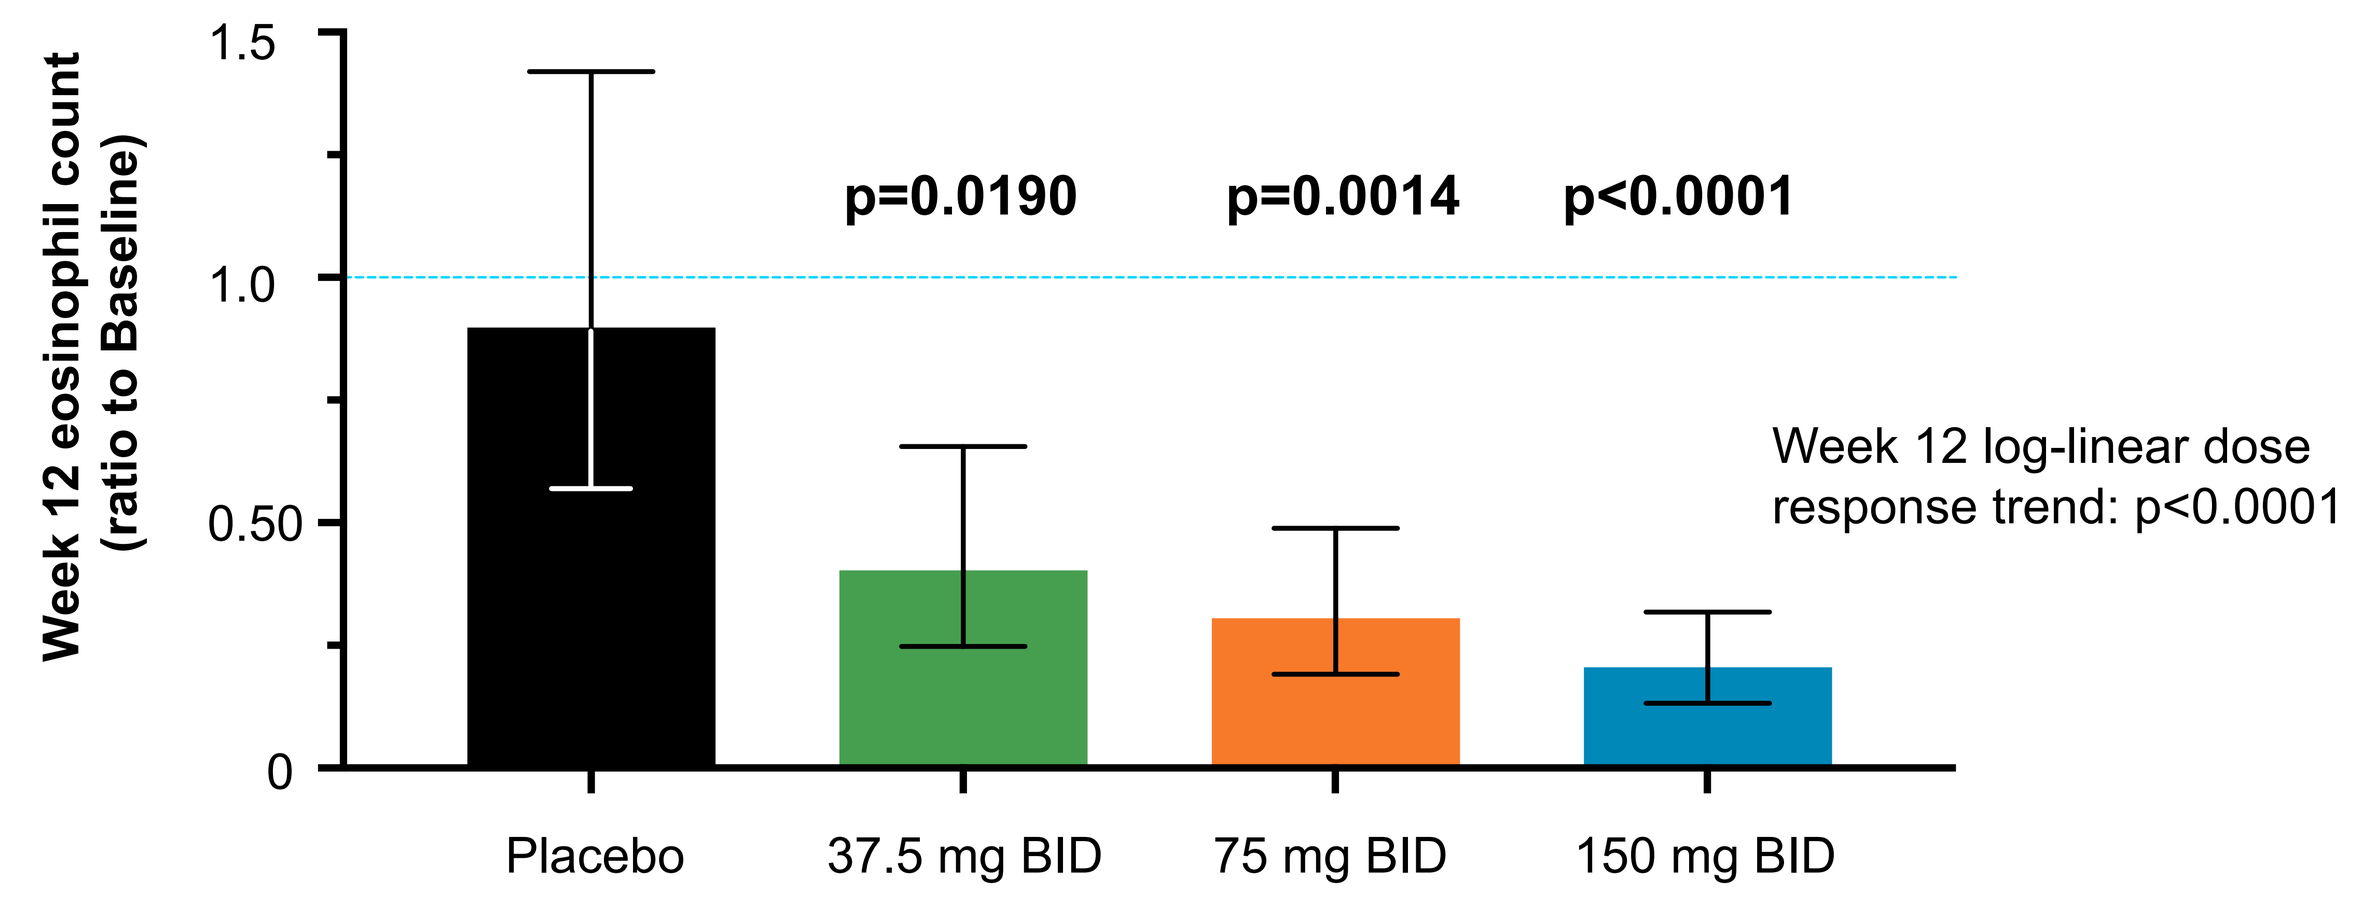

Week 12 eosinophil count (ratio to Baseline)
1.5
1.0
0.50
0
p=0.0190
p=0.0014
p<0.0001
Week 12 log-linear dose response trend: p<0.0001
Placebo
37.5 mg BID
75 mg BID
150 mg BID

Fig. 2: Week 12 absolute eosinophil count ratio to baseline, least square mean using a mixed-effects model with repeated measures. Error bars represent 95% confidence intervals, p values shown for each dose arm vs. placebo.

Fig. 3 Pre-BD FEV_1, change from baseline

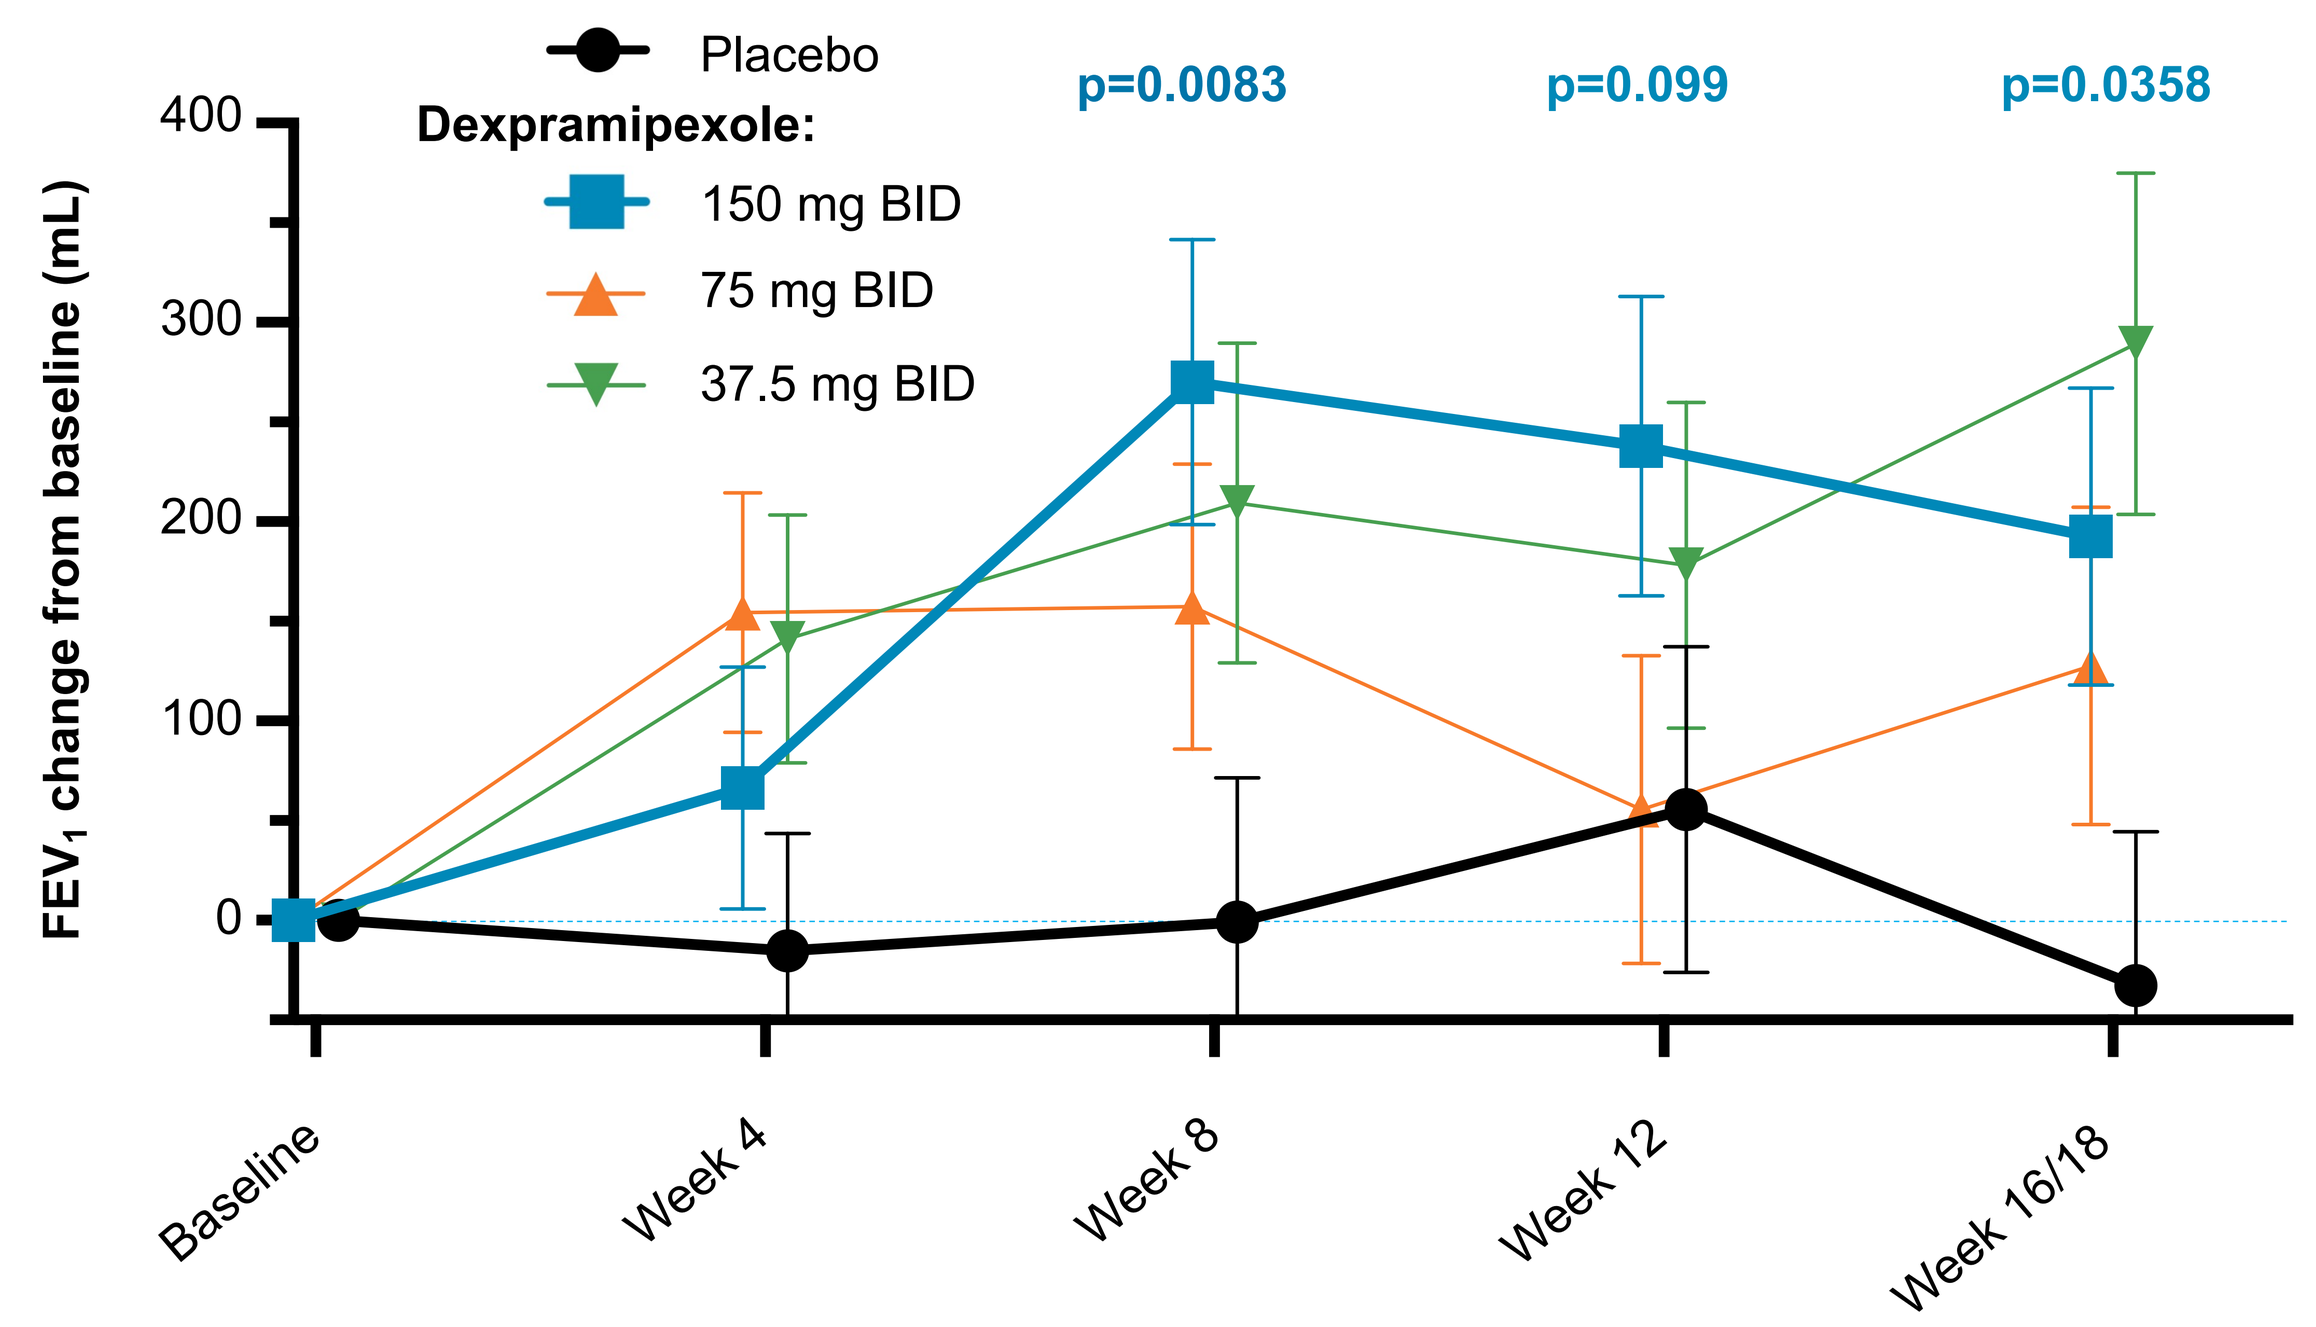

Placebo
Dexpramipexole:
150 mg BID
75 mg BID
37.5 mg BID
p=0.0083
p=0.099
p=0.0358
FEV1 change from baseline (mL)
400
300
200
100
0
Baseline
Week 4
Week 8
Week 12
Week 16/18

Fig. 3: Pre-BD FEV_1 least square mean change from baseline over study visits, using a mixed-effects model with repeated measures. Error bars represent standard error, p values shown for 150 mg BID arm vs. placebo.

Fig. 4 ΔFEV_1 is correlated to Δeosinophils

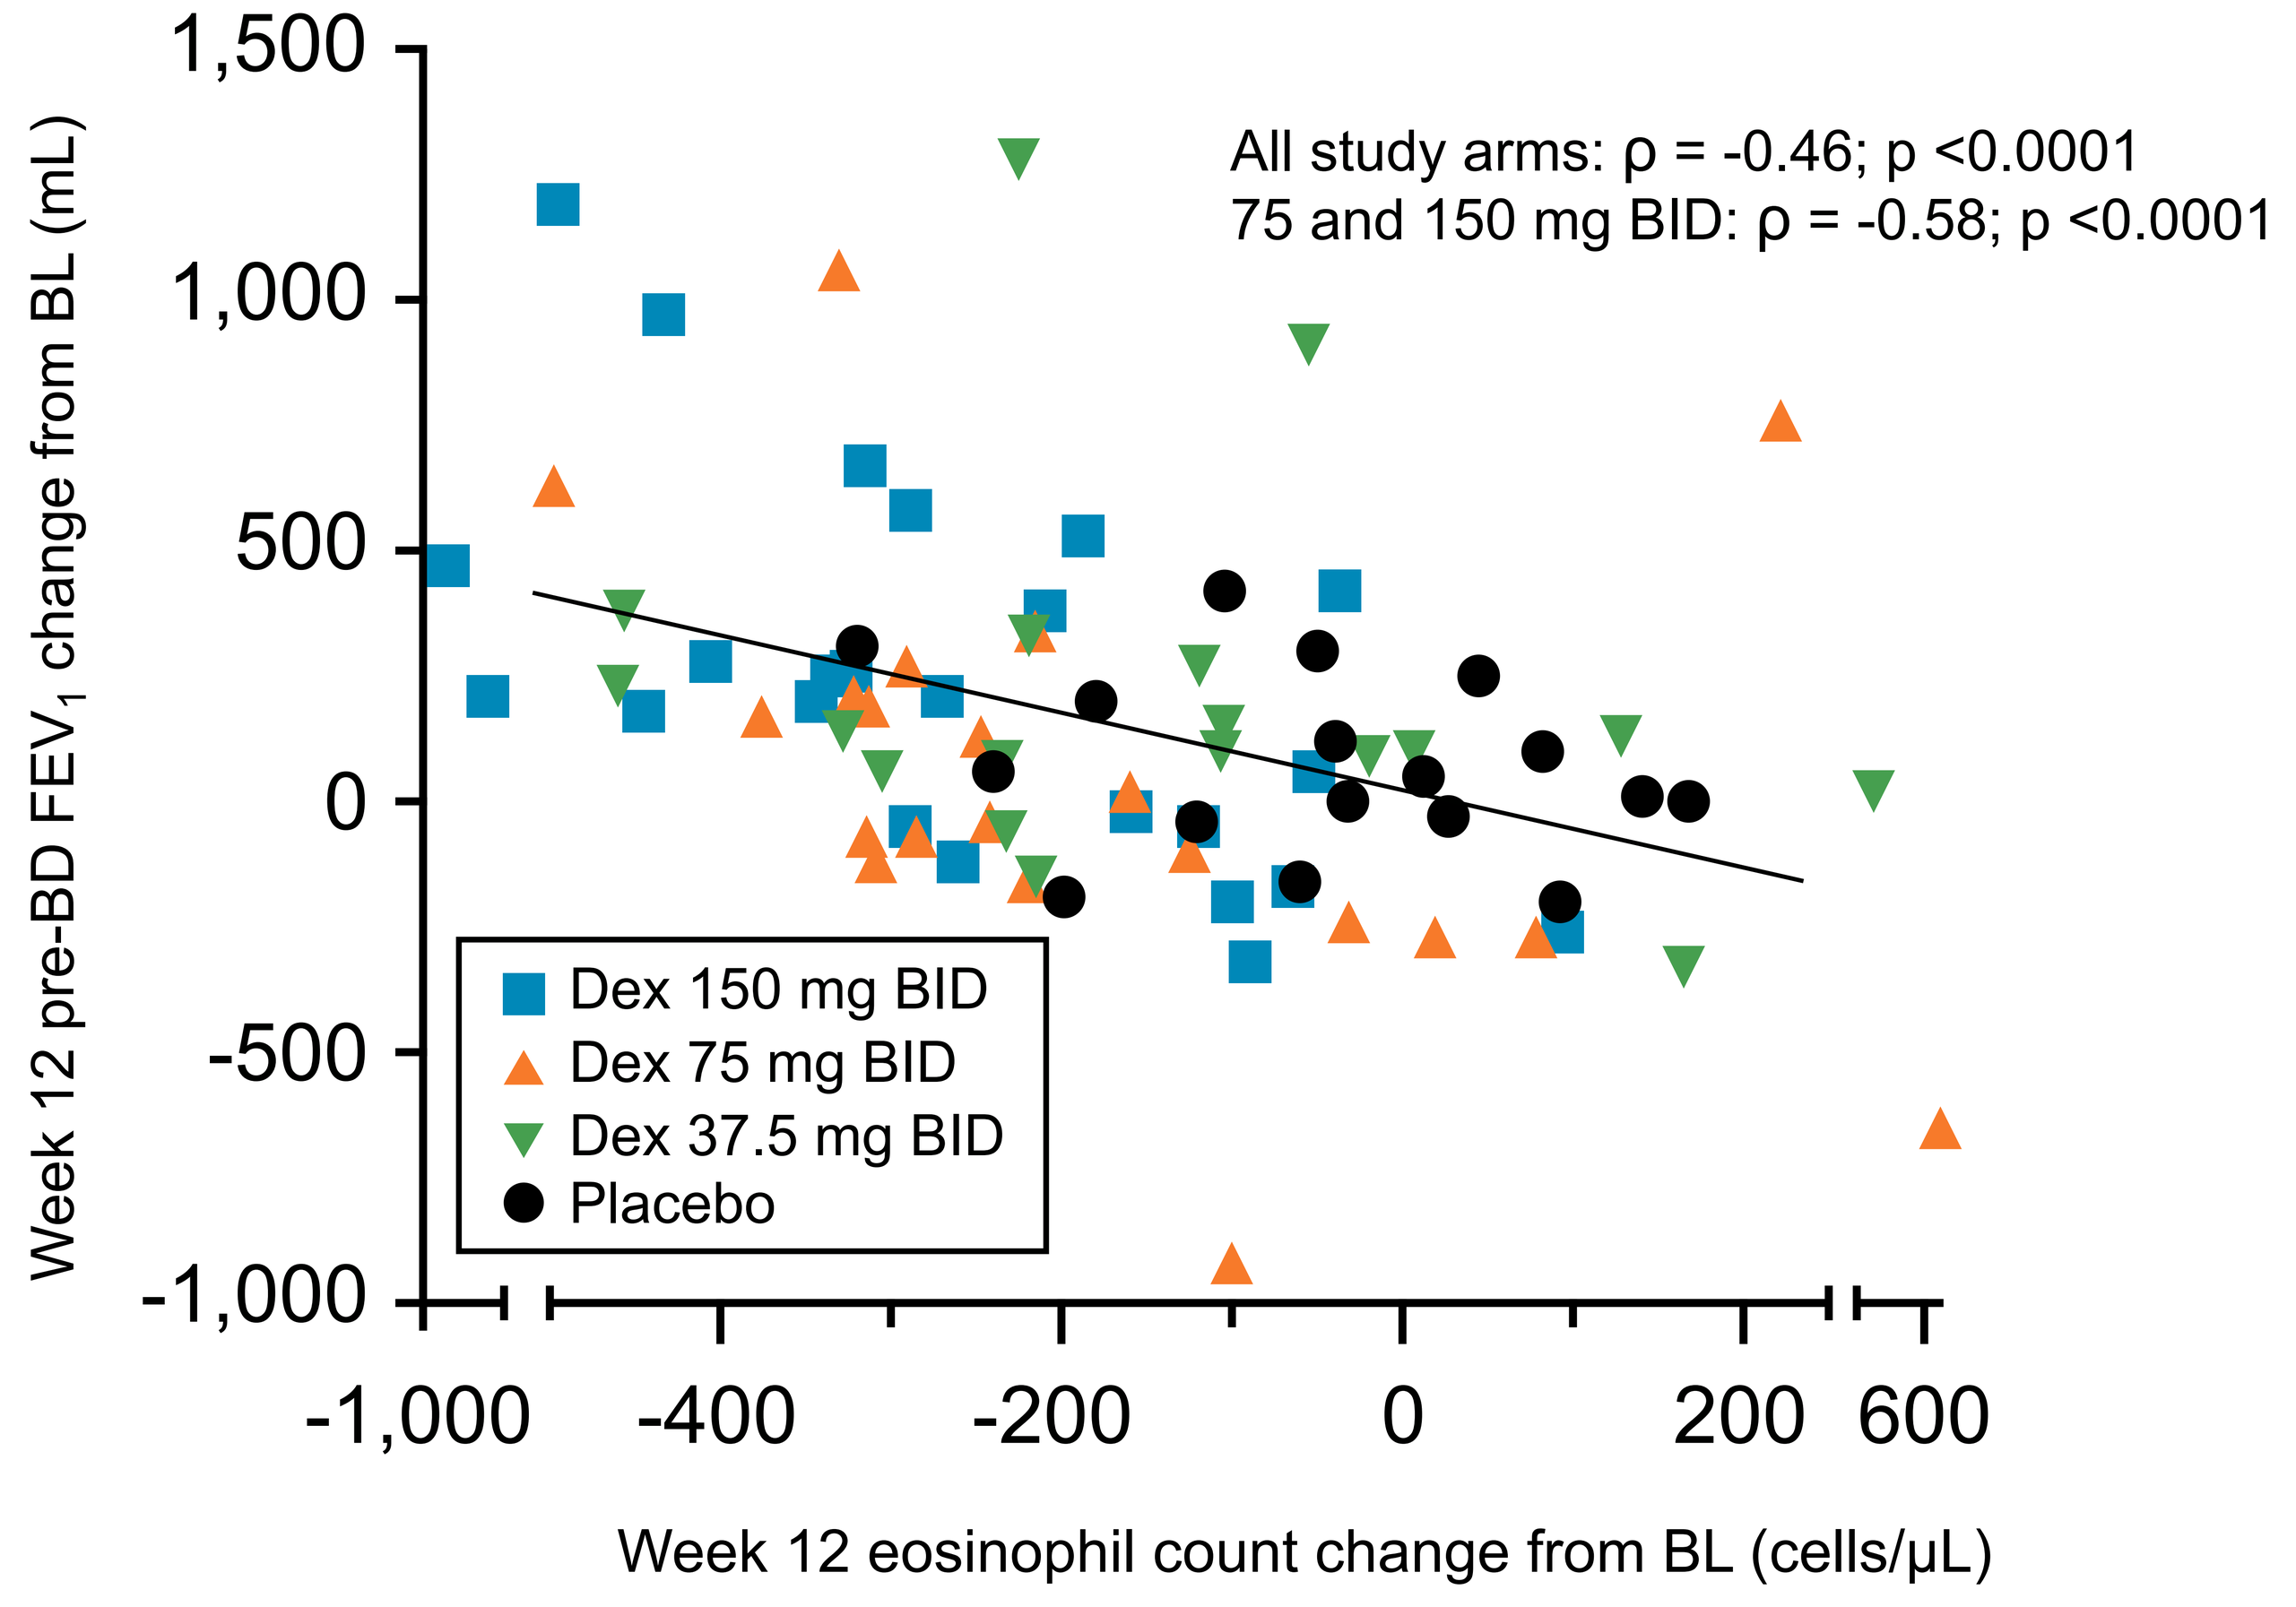

All study arms: ρ = -0.46; p <0.0001
75 and 150 mg BID: ρ = -0.58; p <0.0001
Week 12 pre-BD FEV1 change from BL (mL)
1,500
1,000
500
0
-500
-1,000
Dex 150 mg BID
Dex 75 mg BID
Dex 37.5 mg BID
Placebo
-1,000
-400
-200
0
200
600
Week 12 eosinophil count change from BL (cells/μL)

Fig. 4: Week 12 AEC change from baseline vs. Week 12 pre-BD FEV_1 change from baseline. Each symbol represents a unique subject per the key shown. Spearman correlation coefficients shown for the groups listed.

Fig. 5 Nasal eosinophil peroxidase (EPX), ratio to BL

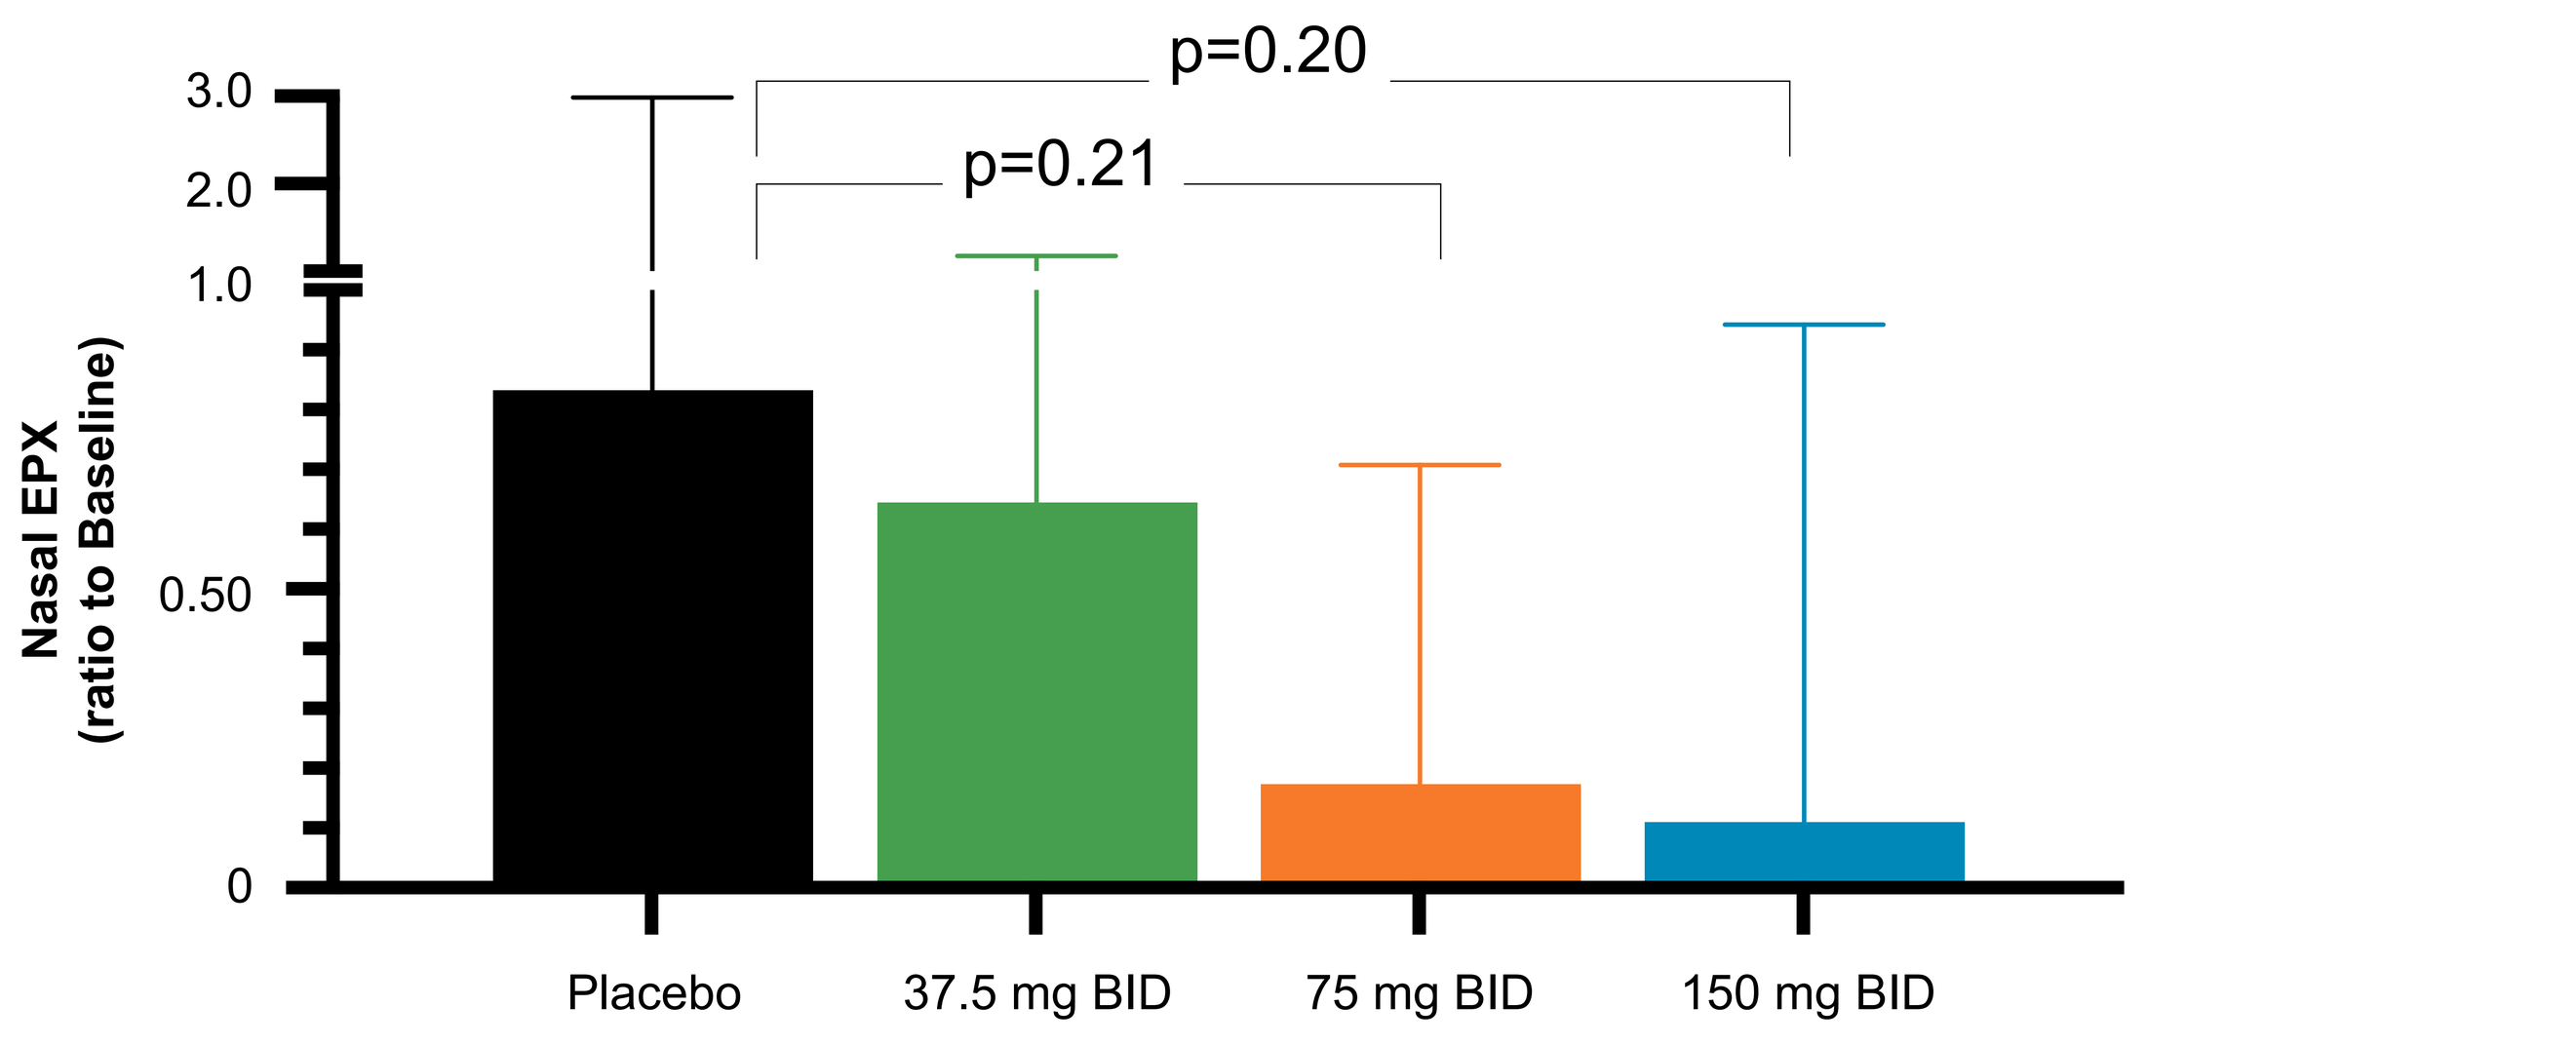

p=0.20
p=0.21
Nasal EPX (ratio to Baseline)
3.0
2.0
1.0
0.50
0
Placebo
37.5 mg BID
75 mg BID
150 mg BID

Fig. 5: Nasal eosinophil peroxidase Week 12 ratio to Baseline for the noted study arms. EPX and total protein were measured in each sample and total protein ratio was used to normalize the EPX for the quantity of sample. Nasal EPX is correlated to sputum eosinophil count and has been identified as a surrogate for lower airway eosinophils.[2]

Fig. 6 FEV_1, FVC, FEV_1/FVC change from baseline (BL)

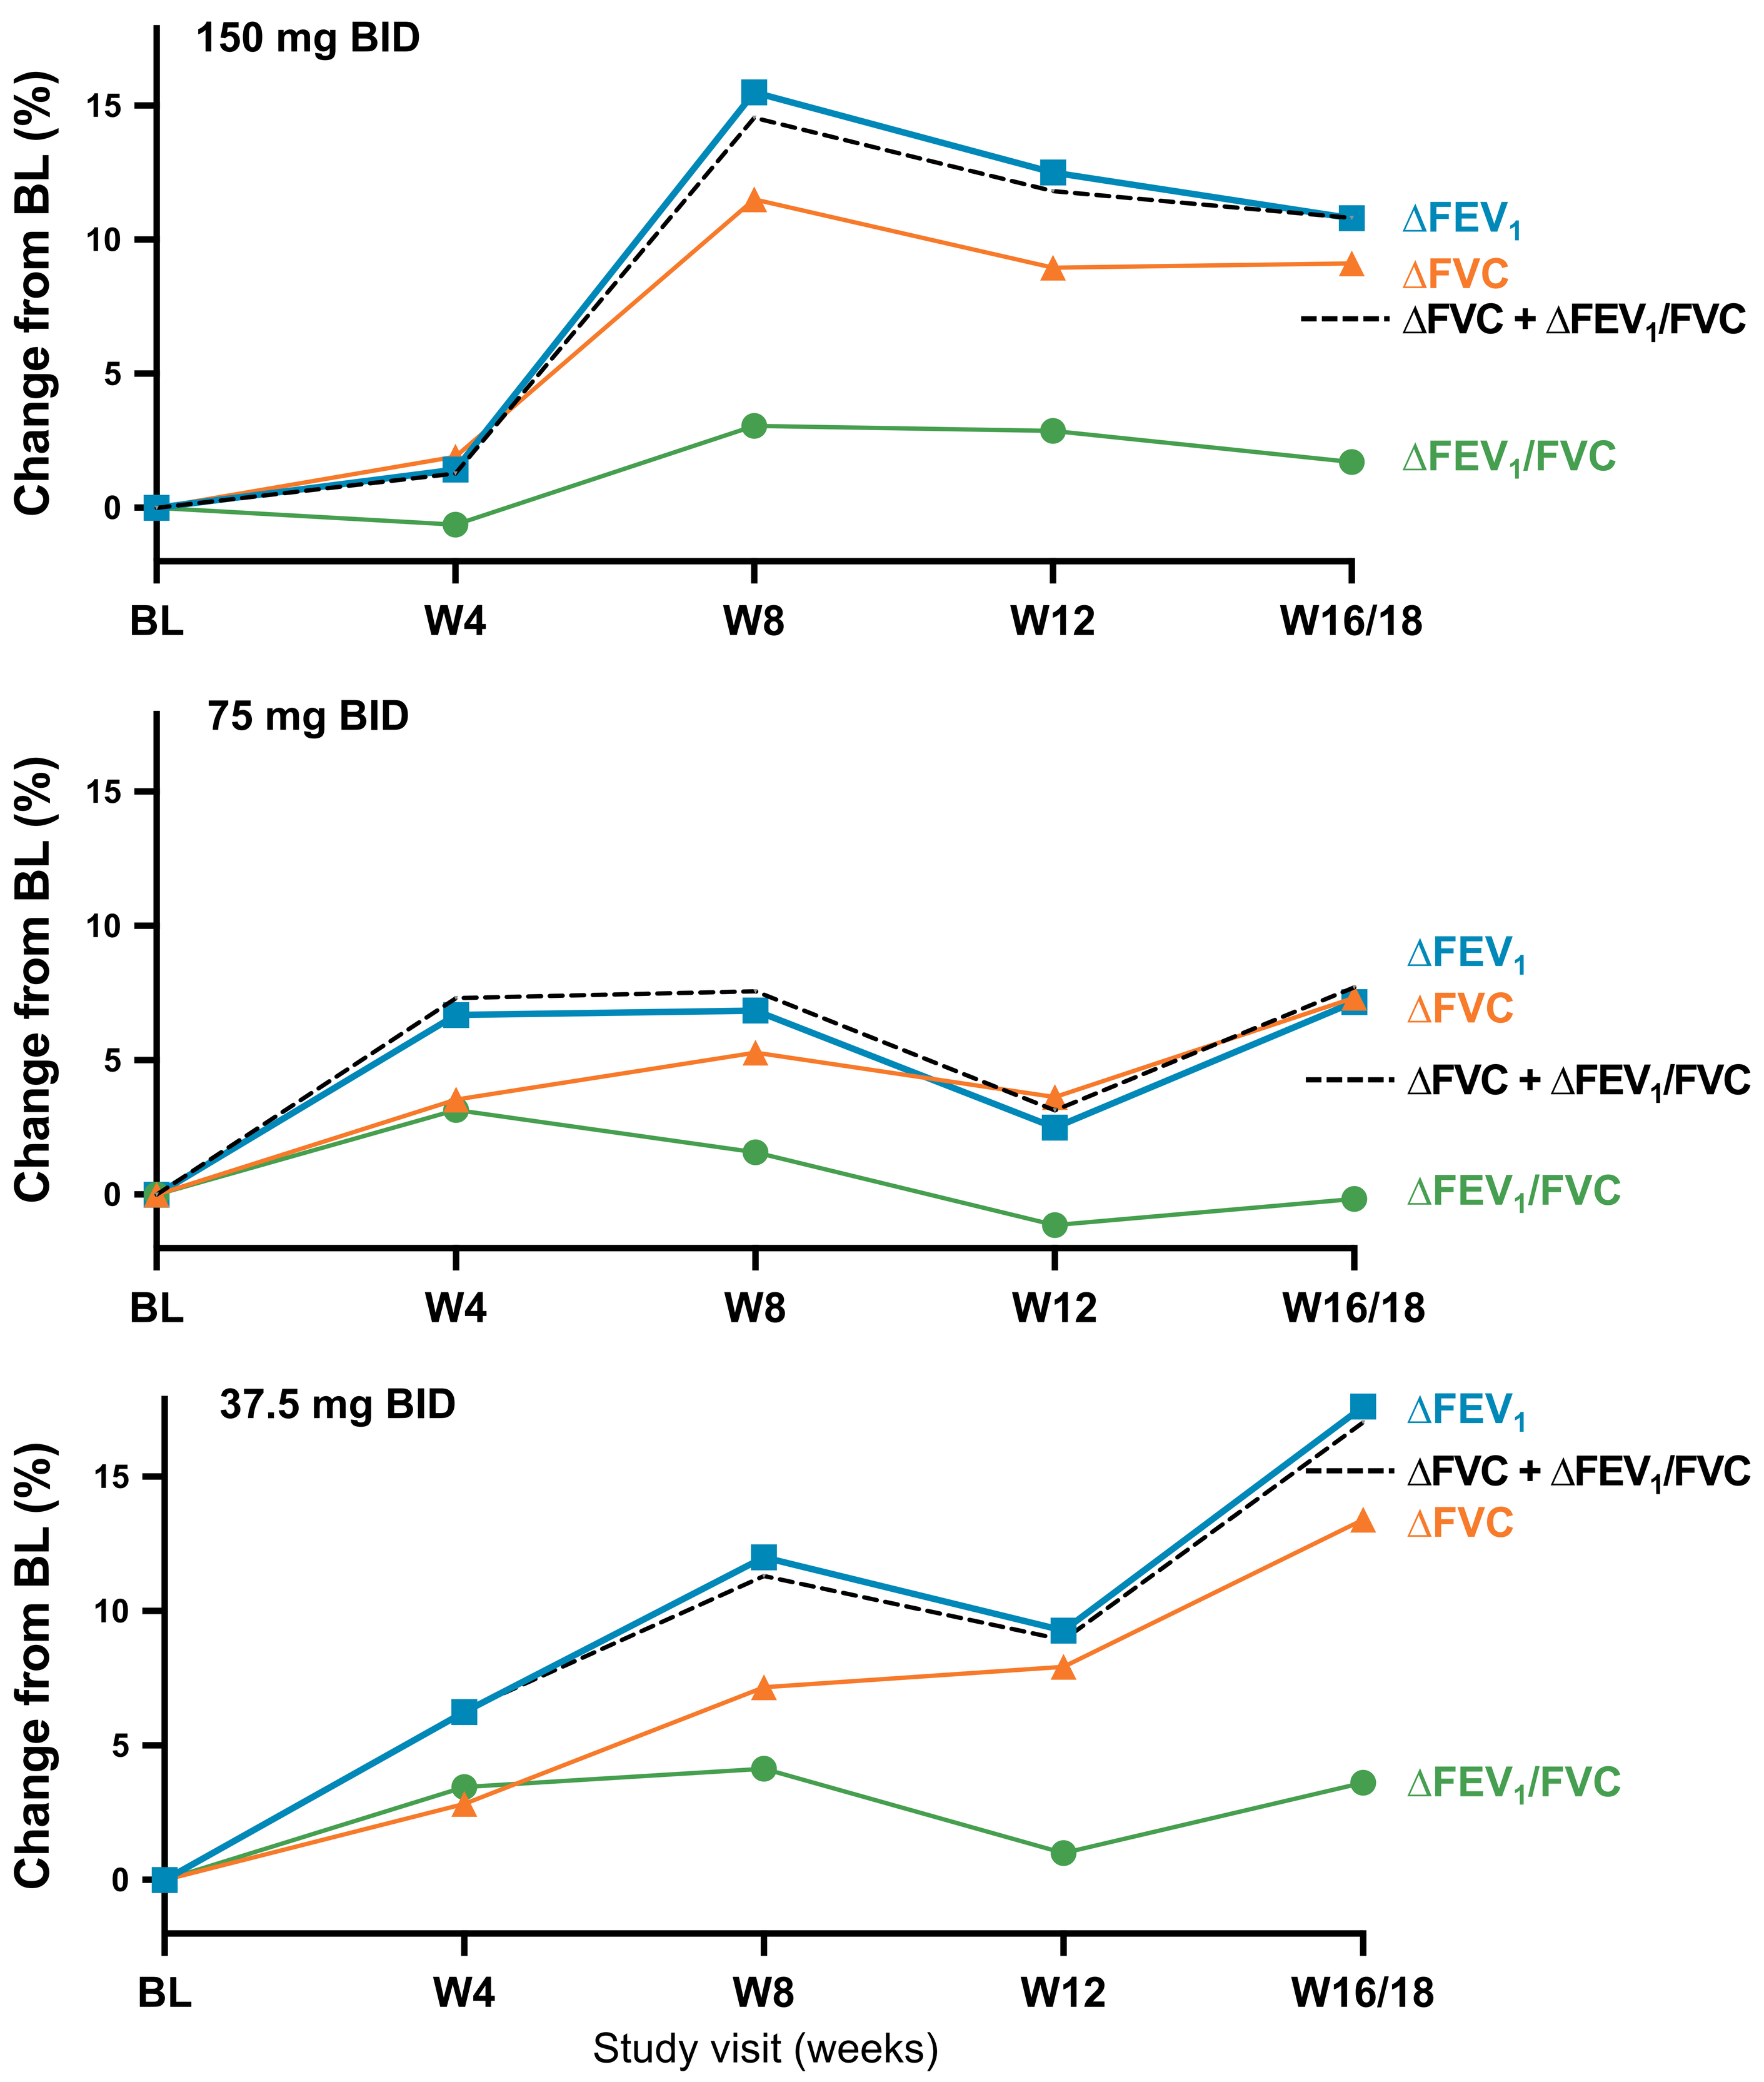

150 mg BID
75 mg BID
37.5 mg BID
Change from BL (%)
15
10
5
0
ΔFEV1
ΔFVC
ΔFVC + ΔFEV1/FVC
ΔFEV1/FVC
BL
W4
W8
W12
W16/18
Study visit (weeks)

Fig. 6: Percent change from baseline for the noted dose groups and spirometric indices.

Conclusions & Discussion

- Dexpramipexole demonstrated significant, dose-dependent lowering of blood eosinophils and nasal EPX, the latter being a surrogate for lower airway eosinophils.
- Dexpramipexole produced clinically meaningful increases in FEV_1 across study arms and time points.
- ΔFEV_1 was correlated with ΔAEC (Fig. 4). These findings clearly demonstrate the link between eosinophil depletion and clinical benefit.
- ΔFVC was the dominant contributor to increases in FEV_1, relative to ΔFEV_1/FVC ratio. This suggests that dexpramipexole, by lowering eosinophils in the airway lumen, may act to decrease mucus plugging and air trapping in small airways, thus leading to predominant improvements in FVC. These findings are consistent with those of Dunican and Fahy, supporting a key role for lower airway eosinophils and EPX in driving mucus plugging and air trapping.[3]
- These findings support continued clinical development of dexpramipexole as a first-in-class oral eosinophil-lowering drug in asthma.

Proposed model:

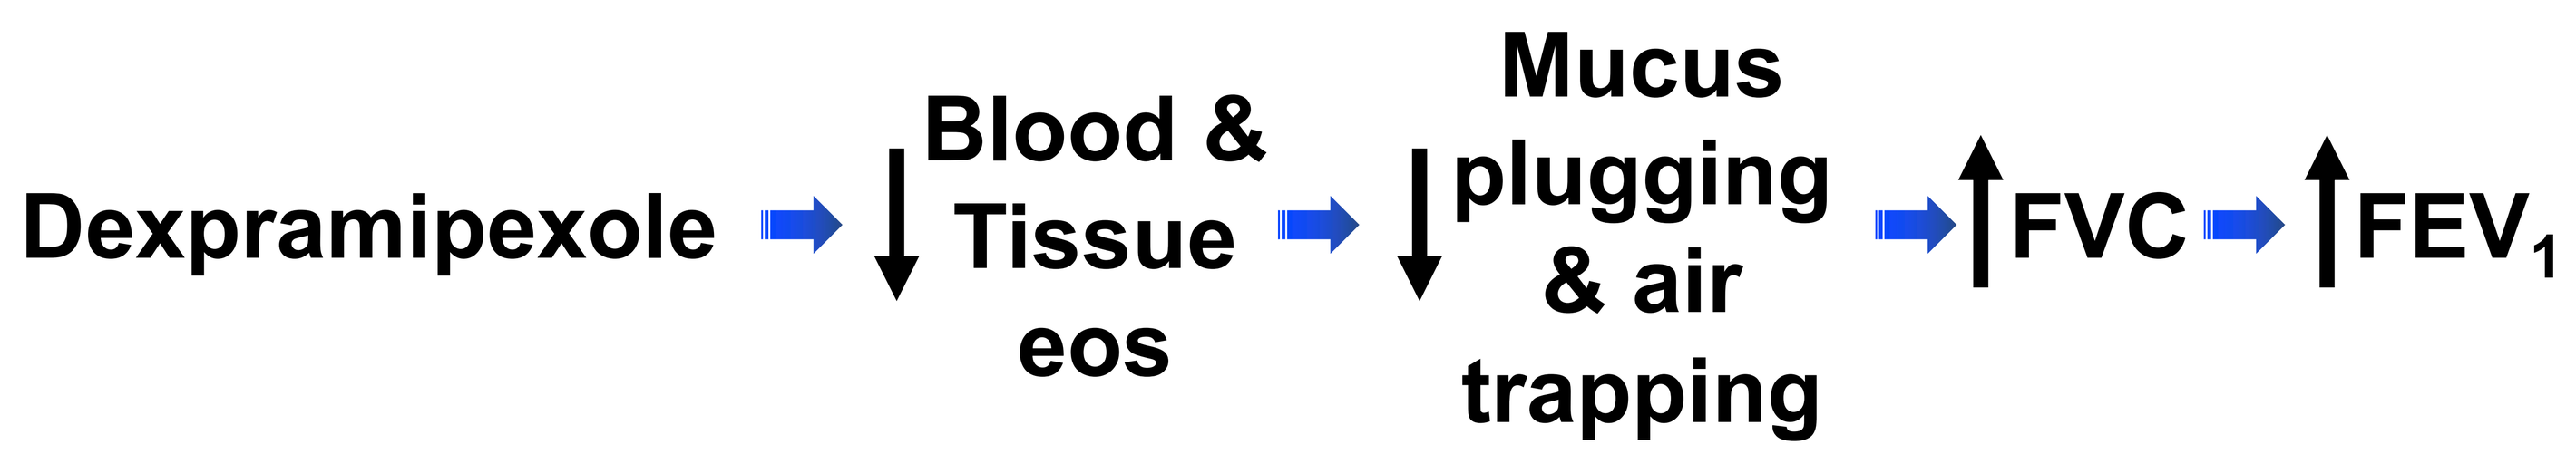

Dexpramipexole
Blood & Tissue eos
Mucus plugging & air trapping
FVC
FEV1

References

1. Sorkness RL, et al. Lung function in adults with stable but severe asthma: air trapping and incomplete reversal of obstruction with bronchodilation. J Appl Physiol 2008 Feb;104(2):394-403.
2. Rank MA, et al. Nasal and pharyngeal eosinophil peroxidase levels in adults with poorly controlled asthma correlate with sputum eosinophilia. Allergy. 2016 Apr;71(4):567-70.
3. Dunican EM, et al. Mucus plugs in patients with asthma linked to eosinophilia and airflow obstruction. J Clin Invest. 2018 Mar 1;128(3):997-1009.

Funding

This study was funded by Knopp Biosciences LLC.